Real-World Genetic Testing Patterns in Prostate Cancer Assessed via Artificial Intelligence: Implications for NCCN Guideline Implementation

Mitchell Singstock, MD[1]; Molly Mendenhall, MBOE, BSN, LSSBB, RN[2]; Suzzette Arnal, PhD[3]; David Waterhouse, MD, MPH, FASCO[2]

1 Department of Internal Medicine, University of Utah Health, Salt Lake City, UT; 2 OHC (Oncology Hematology Care)/US Oncology Network, Cincinnati, OH; 3 The US Oncology Network, The Woodlands, TX

Background:

- Germline and somatic genomic testing in prostate cancer guides both targeted therapy selection and identification of heritable cancer risk.[1,2]
- Although NCCN guidelines define testing eligibility based on disease risk and stage, real-world testing remains suboptimal.[3]
- Population-level data indicate that only one-third of men with metastatic prostate cancer undergo genomic testing.[3]
- As part of a larger effort to improve testing rates, we evaluated three consecutive years of prostate cancer care to quantify testing utilization and assessed AI performance for risk stratification and testing eligibility.

Baseline Measure:

- Conducted a retrospective chart review of patients with advanced prostate cancer treated at a large multi-site community oncology practice between 2023 and 2025.
- Clinical data were manually reviewed to assign disease stage, NCCN risk stratification, eligibility for germline and somatic testing, and presence of testing.
- Structured inputs included: age and PSA at diagnosis, TNM stage, Gleason score, and grade group.
- In 2023, an untrained AI model (ChatGPT 4.0) was provided explicit rules-based NCCN guideline logic (v1.2023) to determine testing eligibility.
- In 2024 and 2025, ChatGPT 5.2 was provided NCCN guideline text (v1.2024 and v1.2025) and asked to infer risk category and testing eligibility without coaching.
- Primary outcomes included observed annual germline and somatic testing rates and AI concordance with manual review.
- Gold standard was manual review of patient charts.

AI Can Follow NCCN Guidelines

Does Your Clinic?

Accuracy of Untrained AI for NCCN - Indicated Genetic Testing

Year	Charts Reviewed	Germline Eligible	Germline Tested	Somatic Eligible*	Somatic Tested	AI Risk Stratification	AI Testing Recommendation Concordance
2023	259	194	54 (28%)	91	24 (26%)	Not assessed	Germline 97%; Somatic 100%
2024	255	195	82 (42%)	86	37 (43%)	235 / 255 (92%)	Germline 100%; Somatic 100%
2025	177	170	107 (63%)	70	56 (80%)	177 / 177 (100%)	Germline 100%; Somatic 100%

**Somatic testing was "Recommended" for patients with metastatic (M1a-c) disease per NCCN 2023-2025*
Germline testing numbers included High Risk, Very High Risk and M1a-c patients.

Three-year retrospective analysis of AI performance and testing utilization in a large community oncology network

Results:

Across 2023–2025, NCCN-indicated germline and somatic testing was inconsistently performed (see table), with significantly increasing rates over three years (Cochran–Armitage trend test: germline $p < 0.001$, somatic $p < 0.001$) attributable to concurrent QI efforts not describe in this study. Testing outside of NCCN guidelines was ordered by clinicians in 10 (4%), 22 (9%), and 31 (18%) patients each year, respectively.

Future Directions:

- AI-based guideline interpretation demonstrated consistently high concordance with manual review and may offer a scalable approach to systematically identify eligible patients using structured data.
- Planned EMR integration will enable real-time testing identification and scaling across diverse practice settings.
- Ongoing analyses seek to clarify which interventions contributed to increased testing rates over the three-year period.

Acknowledgements:

Funding: Pfizer, AstraZeneca

Contact: Mitch.Singstock@hsc.utah.edu; David.Waterhouse1@usoncology.com

Virtual Poster and References: